Presented at the 5th Symposium of the International Cartilage Repair Society, Gent, Belgium. May 26–29, 2004.

La Tour RÉSEAU DE SOINS

Effect of intra-articular sodium hyaluronate (Ostenil®) on improving the quality of life and delaying surgery in patients indicated for total knee replacement. An open, pilot, phase III study

B Mathies MD,[1] J Berger PhD,[2] C Siegfried[2] & R Gurny PhD[2]

[1]Orthopaedic Department, Hôpital de La Tour, 1217 Meyrin, Switzerland;
[2]Department of Pharmaceutical Technology and Biopharmaceutics, School of Pharmaceutical Sciences, University of Geneva, Switzerland.

Correspondence:
Department of Knee Surgery
Hôpital de La Tour
Av. J.-D. Maillard 3
CH-1217 Meyrin
Geneva, Switzerland
Tel: +41 (0)22 719 6677
Fax : +41 (0)22 719 6566
e-mail: burkhard.mathies@latour.ch

Poster number: 397

Introduction

- Viscosupplementation[1] using intra-articular (i.a.) hyaluronic acid (HA) is a recommended treatment option in the management of osteoarthritis (OA) of the knee.[2,3]
- The symptomatic benefits of i.a. treatment with HA have been demonstrated in several studies in patients with knee OA, with beneficial effects lasting between 6 months[4,5] and 1 year after the administration of one i.a. injection of HA per week for five consecutive injections.[6]
- The long-term benefits of HA have been attributed to an improvement in the viscoelastic properties of the synovial fluid.[1]
- We therefore carried out a study in patients with painful advanced knee OA, who were candidates for total knee replacement (TKR) within 3 months, to determine whether a treatment cycle with a sterile isotonic solution of hyaluronan (Ostenil®, TRB Chemedica, Munich, Germany) would delay the time to TKR and improve patients' quality of life.
- We also analysed synovial fluid samples taken from these patients at baseline and at regular intervals after treatment to assess whether viscosupplementation would improve the viscoelastic properties of the synovial fluid.

Patients and methods

Study design: An open, pilot, phase III study.

Inclusion criteria: Male and female patients were included in the study if they showed evidence of painful advanced knee OA, were candidates to receive TKR within 3 months (based on Kellgren–Lawrence scale and severe clinical signs, i.e. WOMAC scores and pain on walking 20 m without support, assessed using the 100 mm Scott–Huskisson visual analogue scale, VAS) and required continuous treatment with non-steroidal anti-inflammatory drugs (NSAIDs). All patients were required to provide signed informed consent.

Exclusion criteria (abbreviated): Factors leading to exclusion from the study included:

- accompanying OA of the ipsilateral hip of sufficient severity to interfere with the functional assessment of the knee
- known or suspected infection of the affected joint
- painful knee conditions other than OA, such as Sudeck's atrophy, synovial pathologies, rheumatoid arthritis or other rheumatoid conditions
- severe obesity (BMI >40)
- treatment with SYSADOAs (symptomatic slow-acting drugs in OA) or i.a. corticosteroid within the 3 months prior to the study start.

Test product: Ostenil® was supplied as a sterile, ready-to-use, pre-filled syringe containing 20 mg/2 ml of highly purified sodium hyaluronate obtained by bacterial fermentation.

Evaluation parameters: The primary efficacy criteria were pain on walking 20 m without support, assessed using the VAS, and pain using the WOMAC Index section A. The secondary efficacy criteria included joint stiffness and function (WOMAC Index sections B and C, respectively), physical examination of the knee, escape medication consumption, quality of life (SF-36 health survey), viscous/elastic moduli of the synovial fluid and efficacy judgements by the patients and the investigator.

Methods: As continuous analgesic or anti-inflammatory medication was an inclusion criterion, no wash-out period was necessary prior to study start. Efficacy parameters were assessed and synovial fluid collected prior to i.a. injection of Ostenil® on Days 0, 7, 14, 21 and 28 (i.e. Visit 1 to Visit 5). Injections were performed in the target knee under standardised conditions using the superolateral approach. When arthrocentesis was required, the amount of synovial fluid obtained was recorded. The treated knee was mobilised immediately after each injection and patients were advised to refrain from strenuous physical activities involving the knee. The synovial fluid samples collected during the study were frozen within 4 hours at -20°C until analysis. Viscoelasticity measurements were made using a RheoStress 1 rheometer (Haake, Karlsruhe, Germany) at a frequency of 1 Hz.

Follow-up visits were scheduled for Days 56 and 84 (Visits 6 and 7). An open visit (Visit 8), which took place during the period from Day 84 up to Month 12, was foreseen to determine the time at which the patient returned for re-treatment because of worsening symptoms (time to re-treatment) or when the patient had TKR (time to TKR). Patients had access to analgesics or NSAIDs as rescue medication and intake was recorded in a patient diary during the study.

Statistical methods

Descriptive statistics were performed on all subjects' characteristics, assessments and measurements at each visit. Inferential statistics were performed on differences from baseline values and after 3 months. The most appropriate methods (Gaussian or non-parametrical techniques) were used.

Results

- A total of 24 patients (average age: 62.5 ± 10.6 years; average weight: 78.9 ± 12.1 kg; 50% female) with painful advanced knee OA [Kellgren–Lawrence grade II (33.3%), III (58.3%) and IV (8.3%)] requiring continuous NSAID treatment and who were candidates for TKR within 3 months were included in the study.
- Three patients dropped out of the study for personal reasons and 21 patients were evaluated. Of these, three underwent TKR between 4.5 and 6 months after the start of treatment while the other 18 did not require TKR in the 12-month period after the start of treatment (end of study). TKR was delayed by a mean of 7.5 ± 2.3 months after a treatment cycle with Ostenil®.
- The primary efficacy parameter of pain in the affected joint on walking 20 m without support decreased significantly (p = 0.0002) from 43.67 ± 14.65 mm (median: 44.0 mm) at baseline to 16.67 ± 16.61 mm (median: 9.0 mm) at Visit 5. This beneficial effect was maintained during the treatment-free follow-up period (Figure 1).

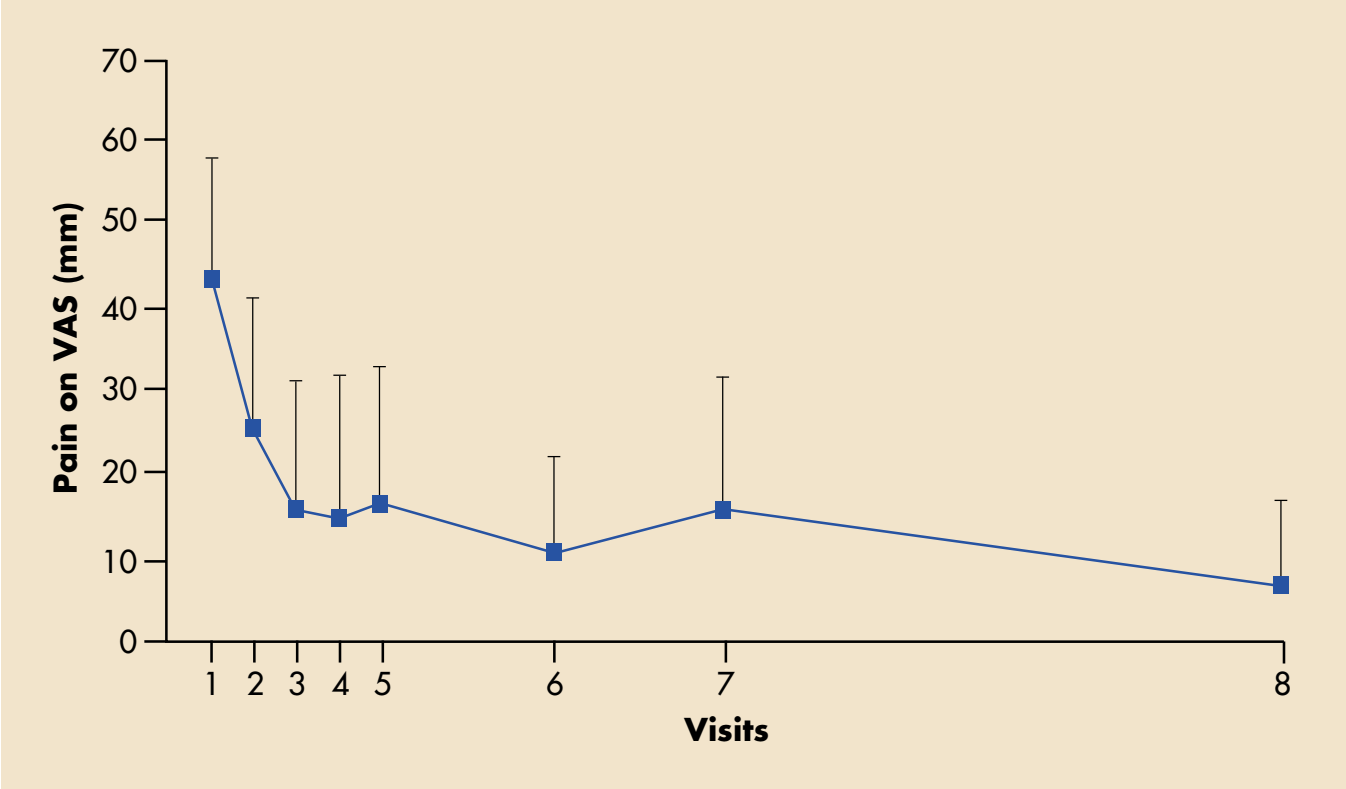
Pain on VAS (mm)
Visits

Figure 1: *Pain on walking 20 m without support, measured on the visual analogue scale (VAS).*

- The same trend was observed for the other primary efficacy criterion, WOMAC A, which showed a significant (p = 0.002) decrease from 6.86 ± 3.02 (median: 7.0) at baseline to 2.43 ± 2.44 (median: 3.0) at Visit 8 (Figure 2).

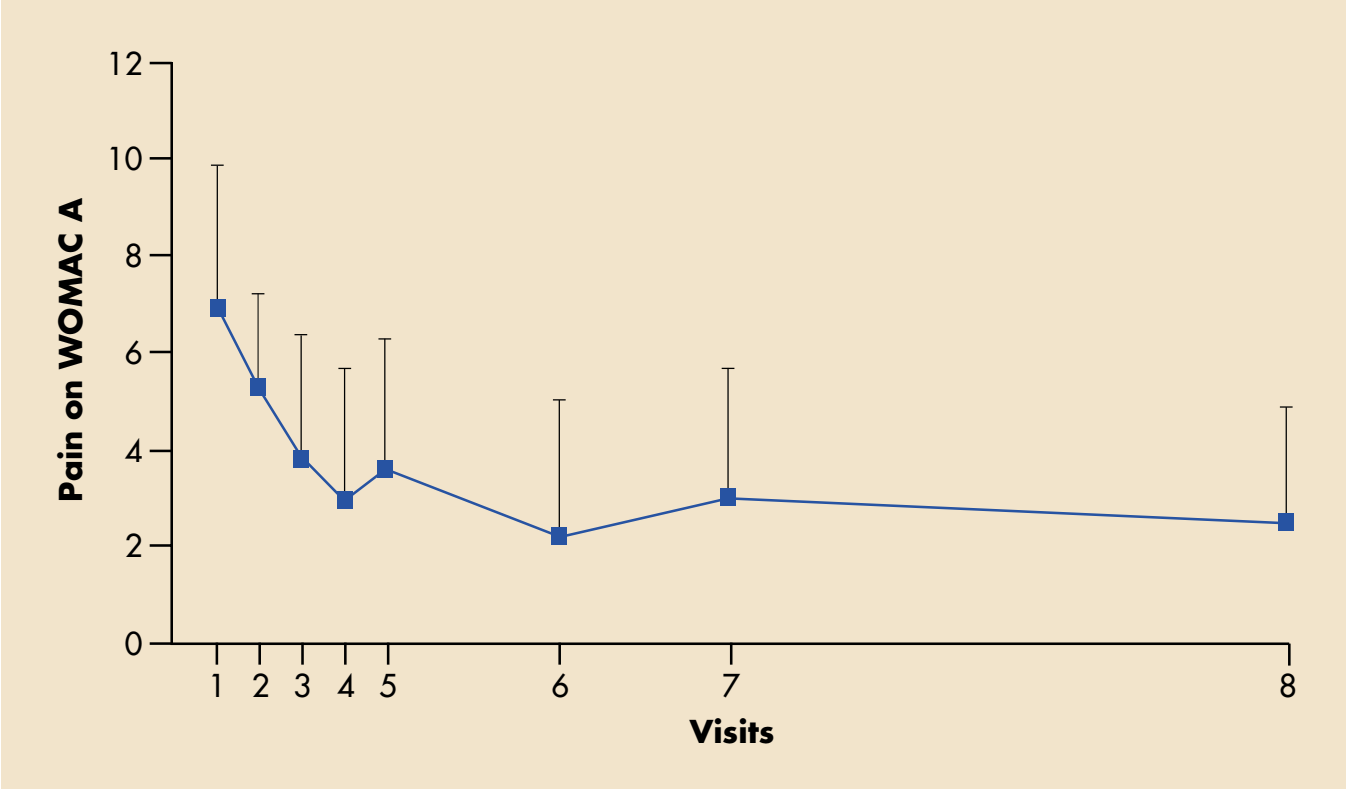
Pain on WOMAC A
Visits

Figure 2: *Pain assessment using WOMAC Index section A.*

- Joint stiffness (WOMAC B) (Figure 3) and impairment (WOMAC C) (Figure 4) improved significantly (p<0.005) from Visit 3 onwards compared with baseline values. WOMAC B showed a significant decrease from 3.24 ± 2.02 (median: 3.0) at baseline to 1.76 ± 1.64 (median: 2.0) at Visit 3. The same trend was observed for WOMAC C, which showed a significant decrease from 21.95 ± 9.55 (median: 21.0) at baseline to 14.29 ± 9.31 (median: 12.0) at Visit 3.

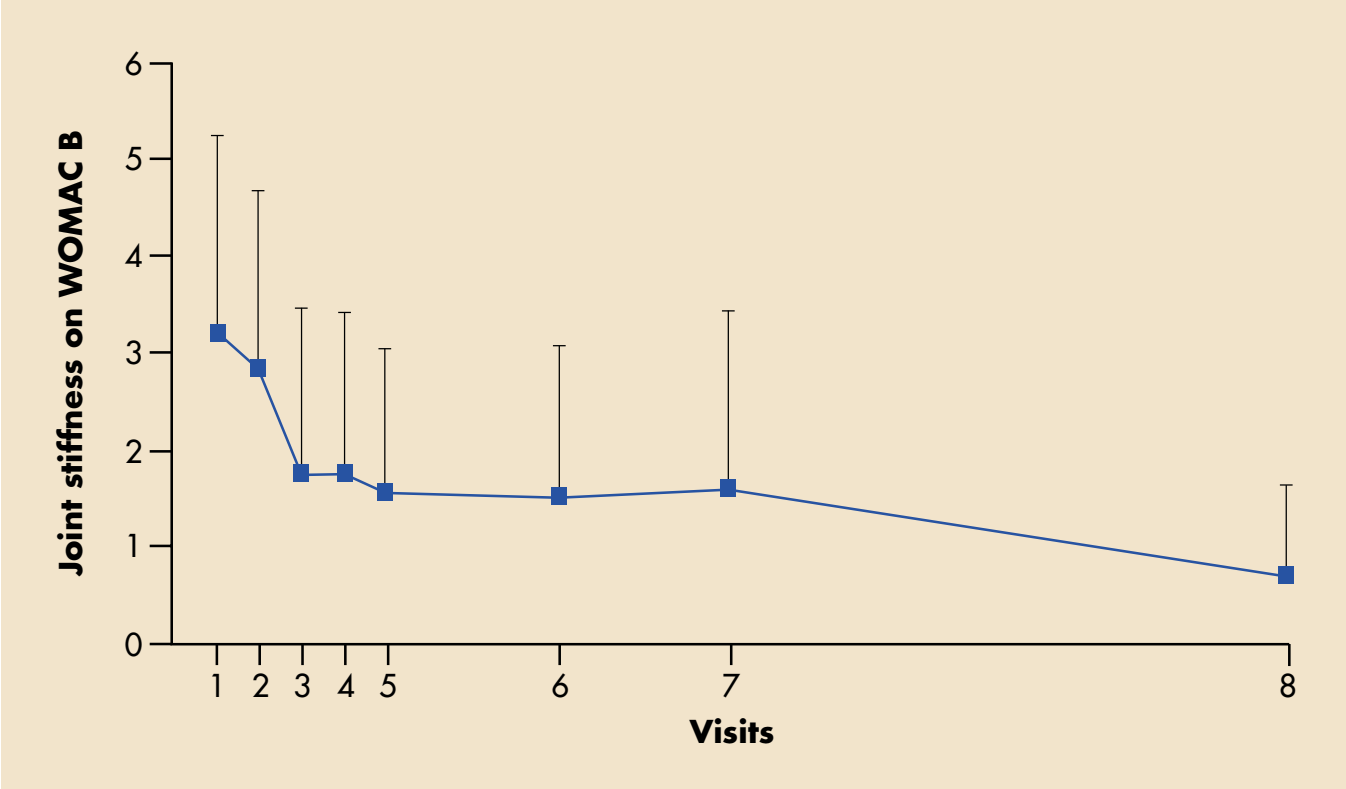
Joint stiffness on WOMAC B
Visits

Figure 3: *Joint stiffness assessed using WOMAC Index section B.*

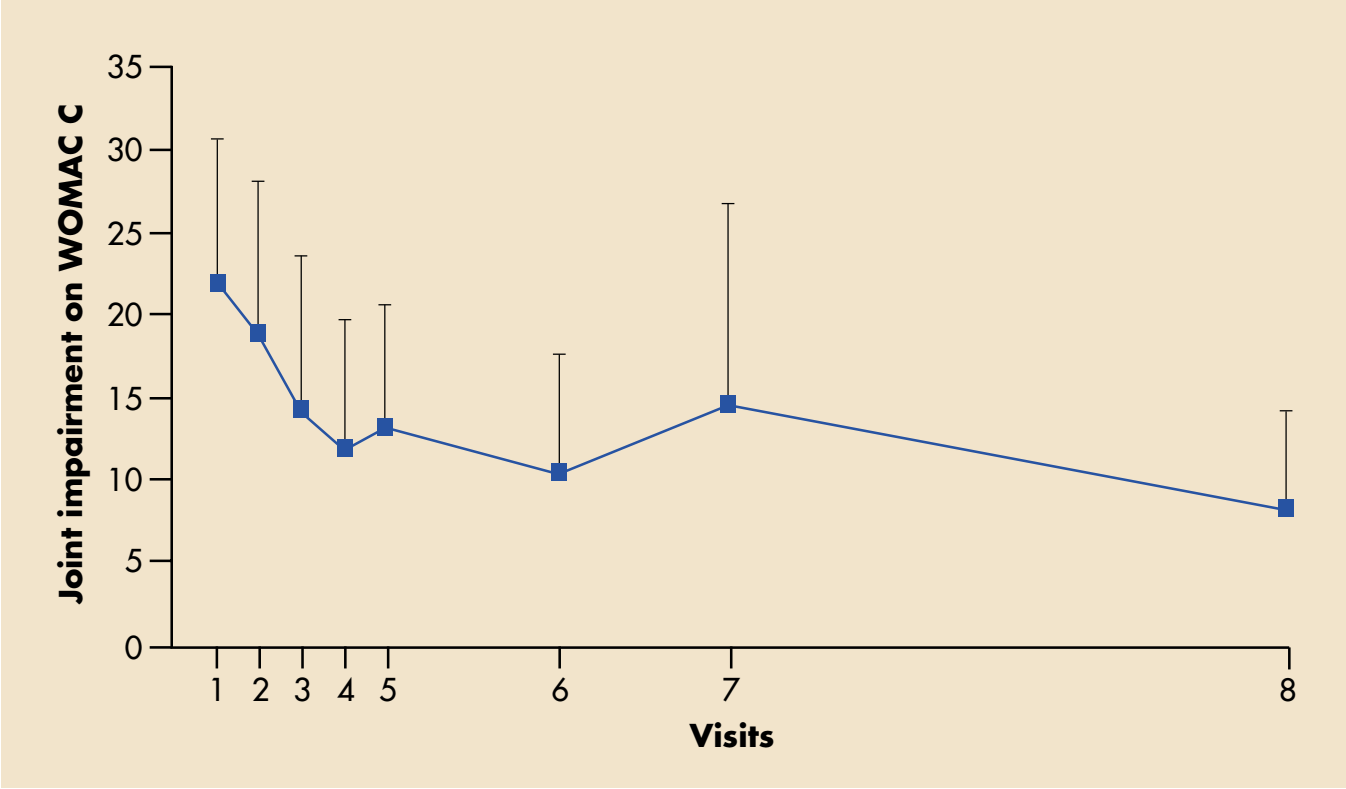
Joint impairment on WOMAC C
Visits

Figure 4: *Joint impairment assessed using WOMAC Index section C.*

- SF-36 improved significantly (p = 0.02) up to Visit 7, with a 22% change in the median score (Figure 5).

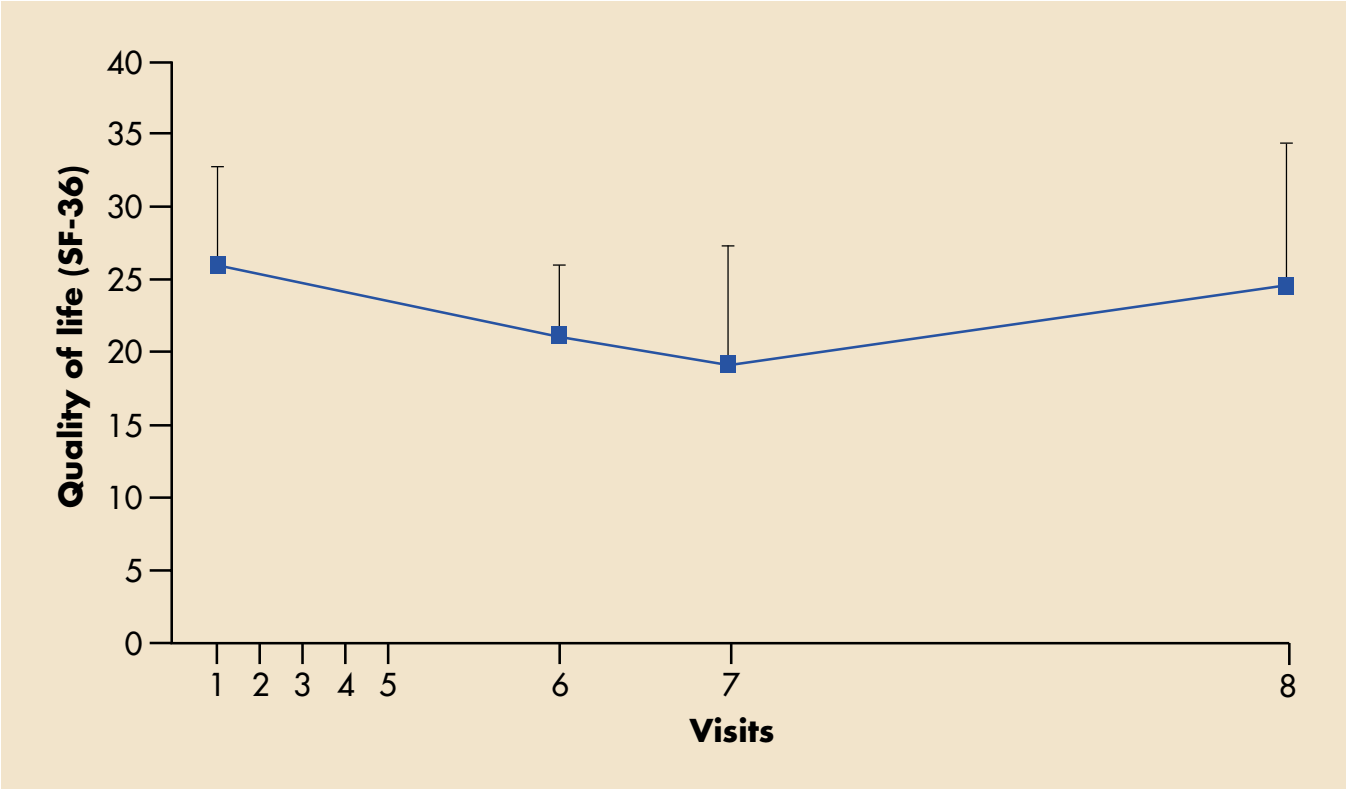
Quality of life (SF-36)
Visits

Figure 5: *Quality of life as measured on the SF-36 health survey.*

- Knee effusion, which was present in 80% of the patients at Visit 1, was found in 57.1% of the patients at Visit 8.
- Consumption of analgesics or NSAIDs did not change during the study period.
- The dynamic elasticity (G′) of the synovial fluid increased from 43.9 ± 8.6 mPa (median: 44.54 mPa) at baseline to 54.0 ± 25.4 mPa (median: 44.16 mPa) at Visit 7 (Figure 6), while dynamic viscosity (G″) increased from 112.1 ± 81.3 mPa (median: 89.83 mPa) at baseline to 171.9 ± 169.6 mPa (median: 104.5 mPa) at Visit 7 (Figure 7). Steady state viscosity (η) increased from 19.5 ± 12.5 mPa.s (median: 16.22 mPa.s) at baseline to 28.9 ± 27.0 mPa.s (median: 18.08 mPa.s) at Visit 7 (Figure 8).

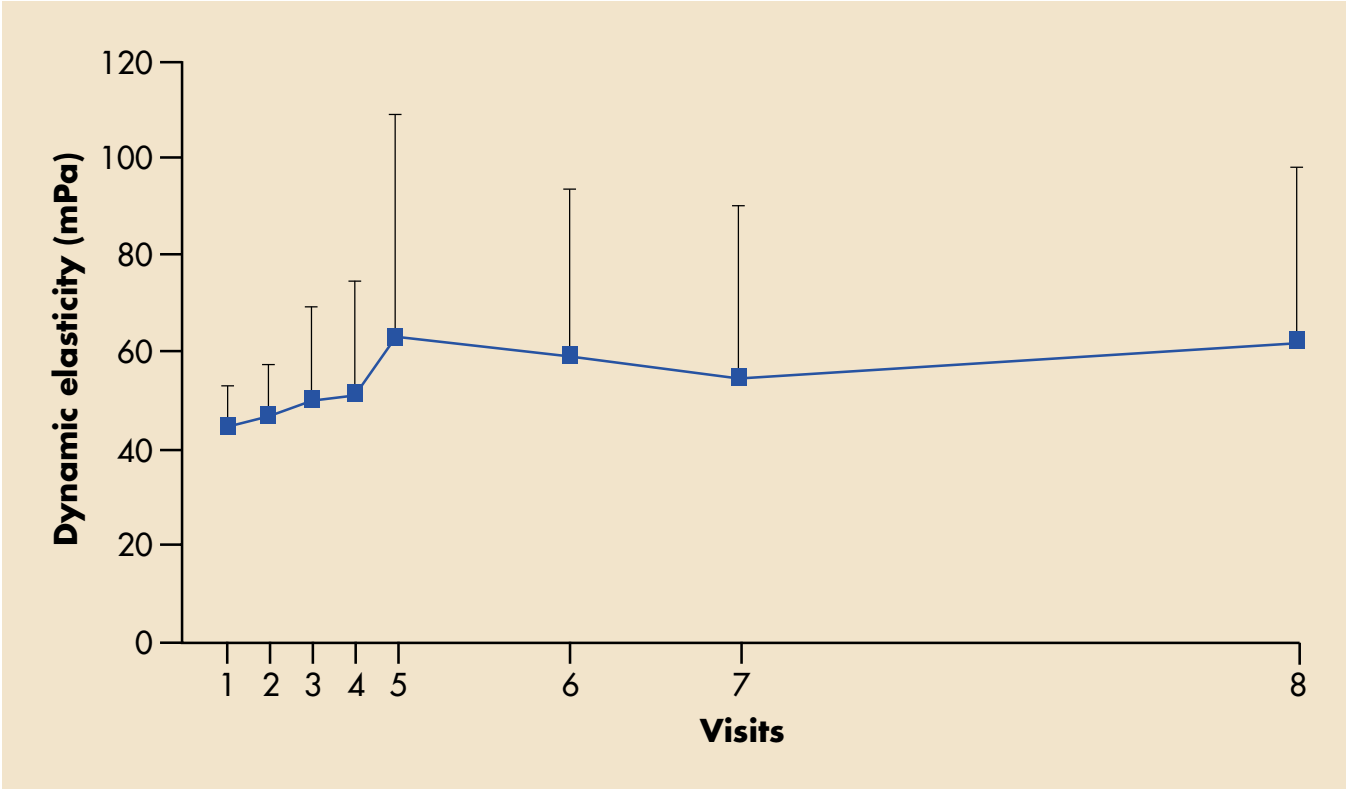
Dynamic elasticity (mPa)
Visits

Figure 6: *Dynamic elasticity of the synovial fluid.*

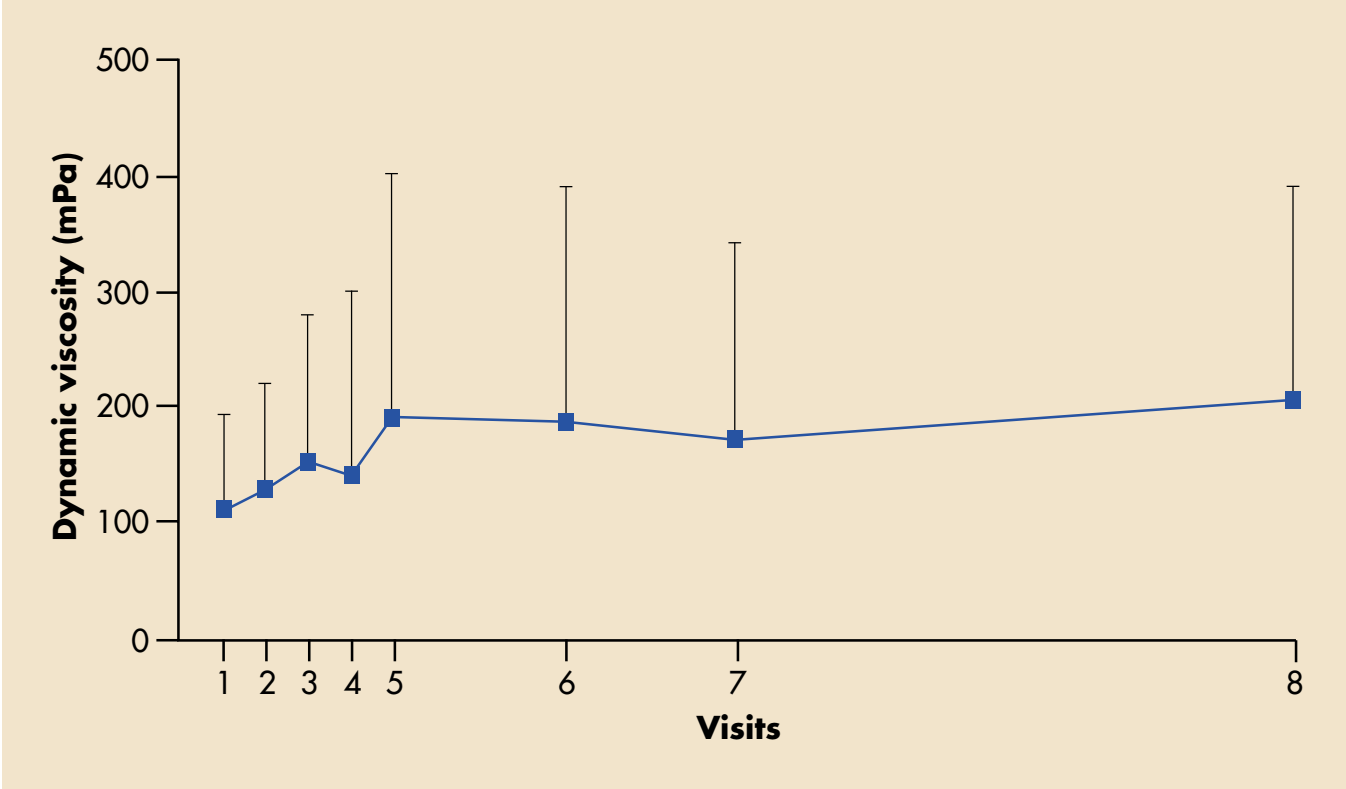
Dynamic viscosity (mPa)
Visits

Figure 7: *Dynamic viscosity of the synovial fluid.*

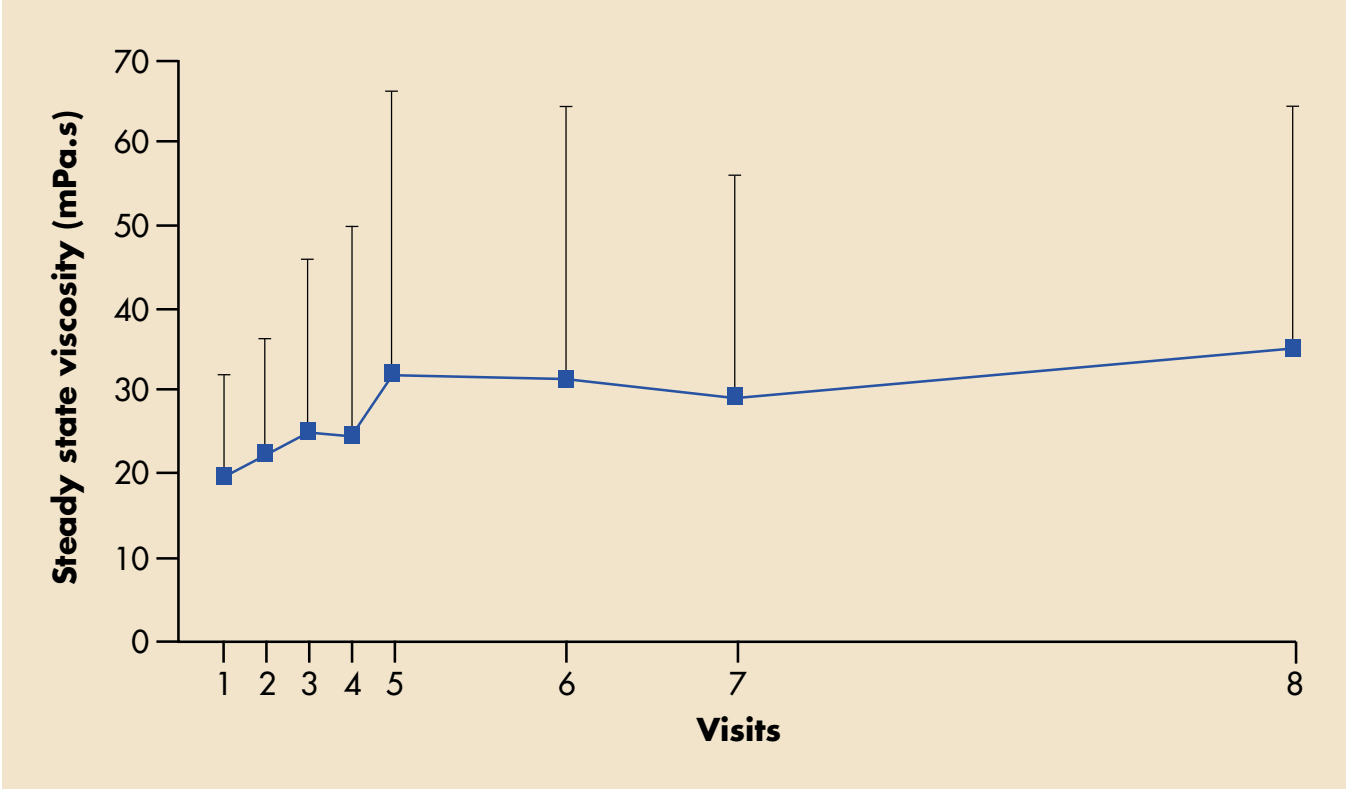
Steady state viscosity (mPa.s)
Visits

Figure 8: *Steady state viscosity of the synovial fluid.*

- Patients judged treatment as 'good' or 'excellent' in 53% of the cases at Visit 6. At the end of the study, 43% of the patients judged the treatment as 'good' or 'excellent'. The efficacy judgements expressed by the investigator showed a similar trend.
- One adverse event (impaired joint function, of moderate intensity, due to effusion occurring one day after the second injection and lasting more than 1 day but resolving spontaneously without sequelae and without the need for other interventions) was reported in one patient.

Conclusions

This open, pilot, phase III study demonstrated that a treatment cycle with Ostenil®:

- was safe and significantly improved symptoms in patients with painful advanced knee OA who were awaiting TKR
- delayed TKR by 4.5 to 6 months in 3 patients and up to 12 months in the other 18 patients
- improved the quality of life of these patients
- improved the viscous and elastic moduli of the synovial fluid, compared with baseline values, which seemed to correspond to the improvement in symptoms.

The relationship between the improvement in clinical signs and the change in viscoelastic properties of the synovial fluid should be further investigated in a larger study.

References

1 Balazs EA, Denlinger JL. Viscosupplementation: A new concept in the treatment of osteoarthritis. *J Rheumatol* 1993; 20(Suppl. 39):3–9.
2 Jordan KM, Arden NK, Doherty M et al. EULAR Recommendations 2003: an evidence based approach to the management of knee osteoarthritis: Report of a Task Force of the Standing Committee for International Clinical Studies Including Therapeutic Trials (ESCISIT). *Ann Rheum Dis* 2003; 62(12):1145–1155.
3 American College of Rheumatology. Recommendations for the medical management of osteoarthritis of the hip and knee: 2000 update. American College of Rheumatology Subcommittee on Osteoarthritis Guidelines. *Arthritis Rheum* 2000; 43(9):1905–1915.
4 Altman RD. A placebo and Naproxen controlled study of intraarticular (IA) hyaluronate (HA) in osteoarthritis (OA) of the knee. *Arthritis Rheum* 1995; 38(9/Suppl.):S240.
5 Huskisson EC, Donnelly S. Hyaluronic acid in the treatment of osteoarthritis of the knee. *Rheumatology* (Oxford) 1999; 38(7):602–607.
6 Kolarz G, Kotz R, Hochmayer I. Long-term benefits and repeated treatment cycles of intra-articular sodium hyaluronate (Hyalgan) in patients with osteoarthritis of the knee. *Semin Arthritis Rheum* 2003; 32(5):310–319.